Mass General Brigham
for Children

NICU nurses' perceptions of partnering with fathers and non-birth parents in the delivery of family-centered care

Carolyn Bleiler, RN PhD

Introduction

- In the U.S., 3.66 million infants were born in 2021. An estimated 9–13% of liveborn infants are admitted to a Neonatal Intensive Care Unit (NICU).
- Family-centered care is the gold standard for nursing care.
- Research indicates that family-centered care is not consistently practiced to its full extent.

Background and significance

- Most research about family-centered care in the NICU has focused on birth mothers.
- There is a paucity of research regarding NICU nurses' perceptions of partnering with fathers or other non-birth parents (NBPs) in family-centered care.

Purpose

- Describe the beliefs and perceptions of NICU nurses throughout the United States regarding partnering with fathers or other non-birth parents in family-centered care.

Research questions

1. **What are NICU nurses' perceptions of partnering with fathers and other non-birth parents in family-centered care in the NICU?**
2. **How do NICU nurses perceive the importance of including fathers or other non-birth parents in family-centered care in the NICU?**
3. **How do NICU nurses involve fathers and other non-birth parents in family-centered care in the NICU?**
4. **Do NICU nurses find meaning in involving fathers or other non-birth parents in family-centered care?**
5. **What are the barriers and facilitators to including fathers and other non-birth parents in family-centered care in the NICU?**

Theoretical framework

- Salutogenesis
- Model of Health: Introduced by Antonovsky (1979)
- View of health: "ease/dis-ease" continuum v. "healthy" or "sick"
 - Main concept is Sense of Coherence
 - Comprehensibility
 - Manageability
 - Meaningfulness

Antonovsky's Sense of Coherence

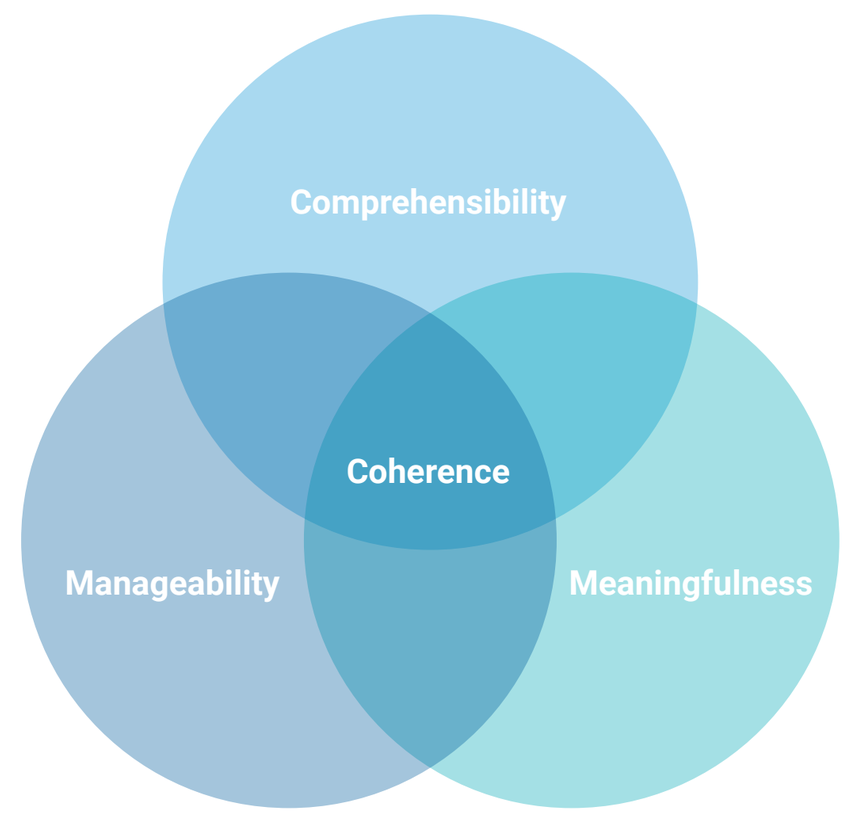
Comprehensibility
Coherence
Manageability
Meaningfulness

Reference: https://sonjablignaut.medium.com/the-value-of-sense-of-coherence-in-organisational-contexts-e4f33dcc265

Qualitative methodology

- Interpretive description
 - Non-categorical, without strict methodological guidance
 - Discover themes & relationships among data, while acknowledging individual discrepancies

Data collection

- Individual semi-structured interviews via Zoom
- Audio-recording
- Verbatim transcription by Rev.com
- Reflexivity journal—reflect on personal beliefs and values, personal preferences and biases

Sample and setting

- NICU nurses currently practicing at the bedside
 - 1 year of bedside NICU experience
- 10 respondents (1 did not meet criteria; 1 did not attend interview)
- Final sample of 8 NICU nurses

Limitations

- Purposive sampling
- Potential bias of researcher
- Potential social desirability bias

Summary/conclusion

- Family-Centered Care is not practiced consistently
- NICU nurses believe it is important to partner with fathers/NBPs in the provision of FCC in the NICU
- Several barriers to partnering with fathers/NBPs were identified
- Understanding NICU nurses' perceptions of partnering with fathers/NBPs can be the impetus for overcoming barriers and designing interventions to improve FCC in the NICU

References

Antonovsky, A. (1996). The salutogenic model as a theory to guide health promotion. Health Promotion International, 11(1), 11-18. https://doi.org//10.1093/heapro/11.1.11.

Centers for Disease Control and Prevention. (n.d.). About Natality 2016-2020 Expanded. http://wonder.cdc.gov/natality-.

Mittelmark, M.B., Sagy SI., Eriksson, M., Bauer, G.F., Pelikan, J.M., Lingstrom, B., & Espnes, G.A. (Eds.). (2017). The Handbook of Salutogenesis. Springer.

National Association of Neonatal Nurses. (2011). Age appropriate care of the premature and critically ill infant: Guideline for practice.

Thorne, S. (2016). Interpretive description: Qualitative research for applied practice (2nd ed.). Rutledge.

Results

1. What are NICU nurses' perceptions of partnering with fathers and other non-birth parents in family-centered care in the NICU?

- **Fathers afraid to touch baby**
- **Fathers seen as secondary to mom**
 - "There is a male presence in a female space"
- **Fathers seen as stressed individuals**
- **Fathers not as present at the bedside as mothers**
- **Families seen as a hindrance**

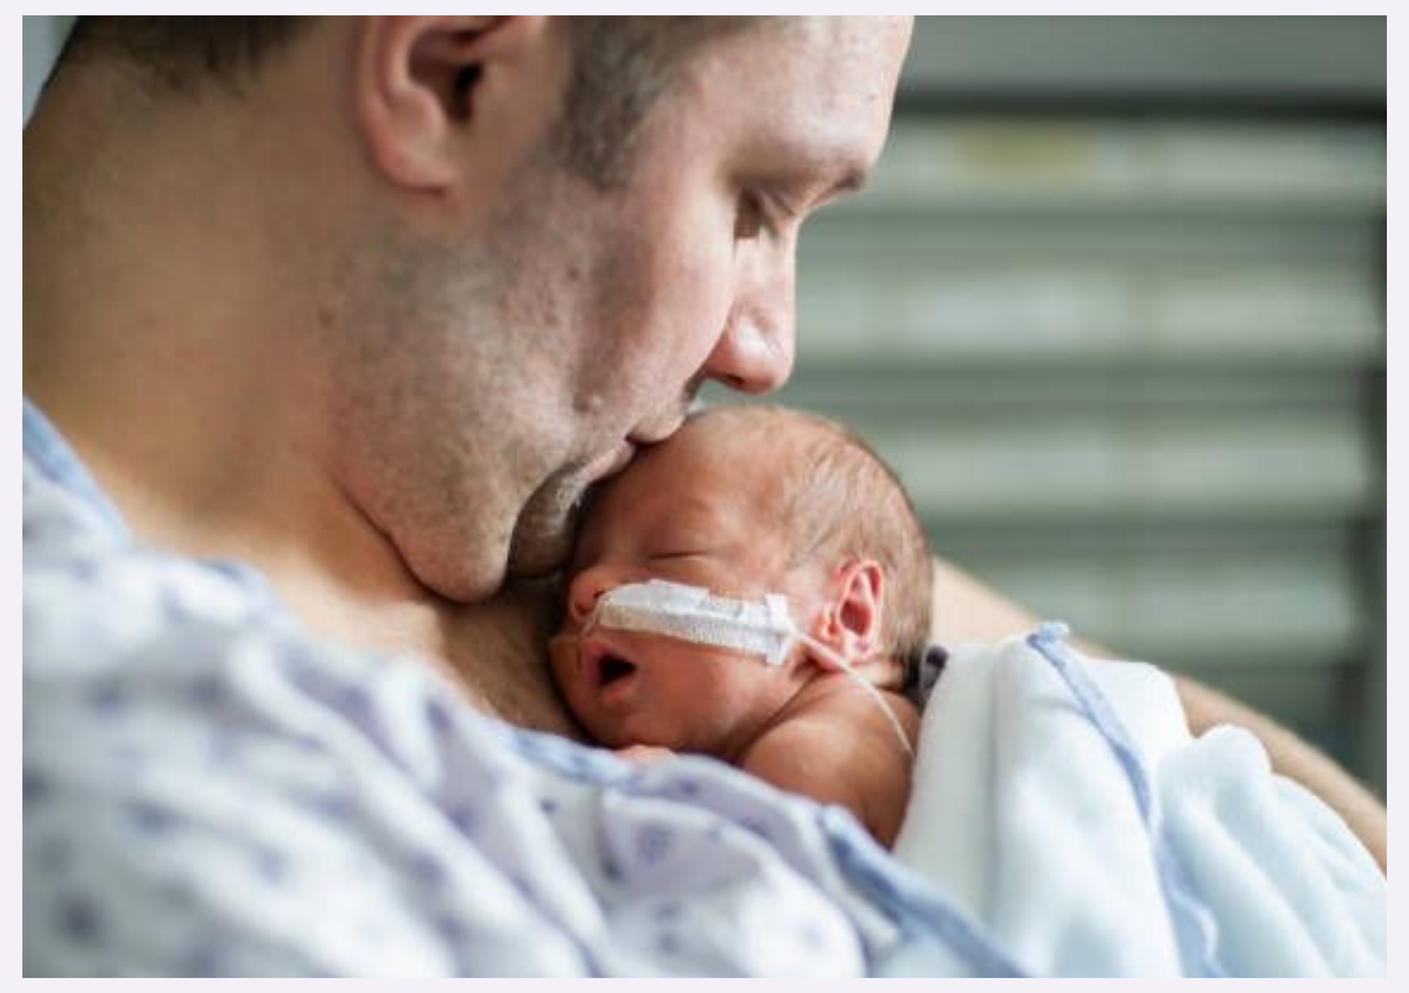

Photo Source: https://www.facebook.com/photo/?fbid=1158234739649076&set=a.418942863578271

2. How do NICU nurses perceive the importance of including fathers or other non-birth parents in family-centered care in the NICU

- **"Fathers are important"**
- **Fathers are "one of the primary caregivers"**

3. How do NICU nurses involve fathers and other non-birth parents in family-centered care in the NICU?

- **Involvement starts upon admission**
- **Some participants report involving fathers/NBPs same as mothers**
 - ". . . you try to encourage the dad to do the other things because he doesn't get to do the breastfeeding part."
- **Some participants revealed involvement depends on how father/NBP presents themselves**
 - "If they don't ask, they don't get" (nurse too busy to make extra effort)
- **Surrogate mother involvement**
 - Each participant expressed that they would defer to parents re: how involved the surrogate mother would be in FCC
- **Same-sex parents**
 - Same-sex parents treated the same as traditional parents
- **Adoptive parents**
 - Overwhelming sentiment: same-sex and/or adoptive parents are treated the same as biological or traditional parents.
 - "It's all about for love of baby"
 - "Regardless of how you're entering this relationship, we're still going to have you definitely part of the team."

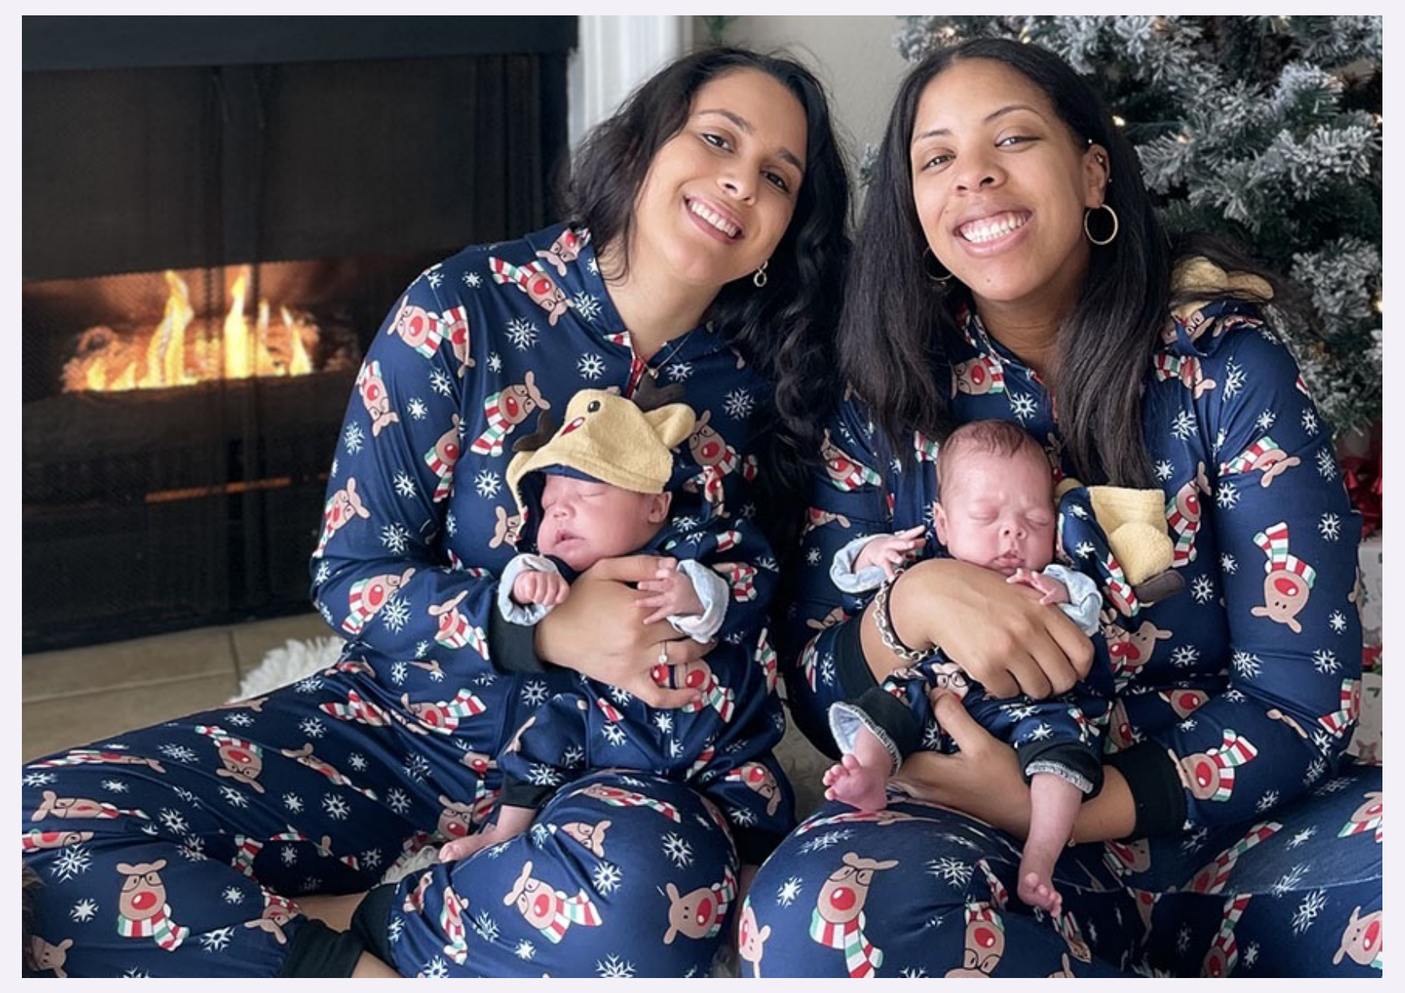

Photo Source: Double the joy: moms bring twins home from NICU | Cedars-Sinai

4. Do NICU nurses find meaning in involving fathers or other non-birth parents in family-centered care?

- **All participants affirmed that involving fathers/NBPs in FCC is important**
 - "I personally like to get the dads to do just as much as the moms."
- **Positive impacts of FCC were identified (parents):**
 - "Establish connection; better outcomes; less burden on mom; infant will have two very supportive parents who do feel comfortable in their relationship and with the care; help infants in development and growth"

5. What are the barriers and facilitators to including fathers and other non-birth parents in family-centered care in the NICU?

- **Lack of nurse's time**
- **Infant safety**
- **Dad's personal barriers; social stigma; cultural barriers**
- **Culture; language barrier**
- **Distractions**
- **NICU environment**
- **Family/social barriers**

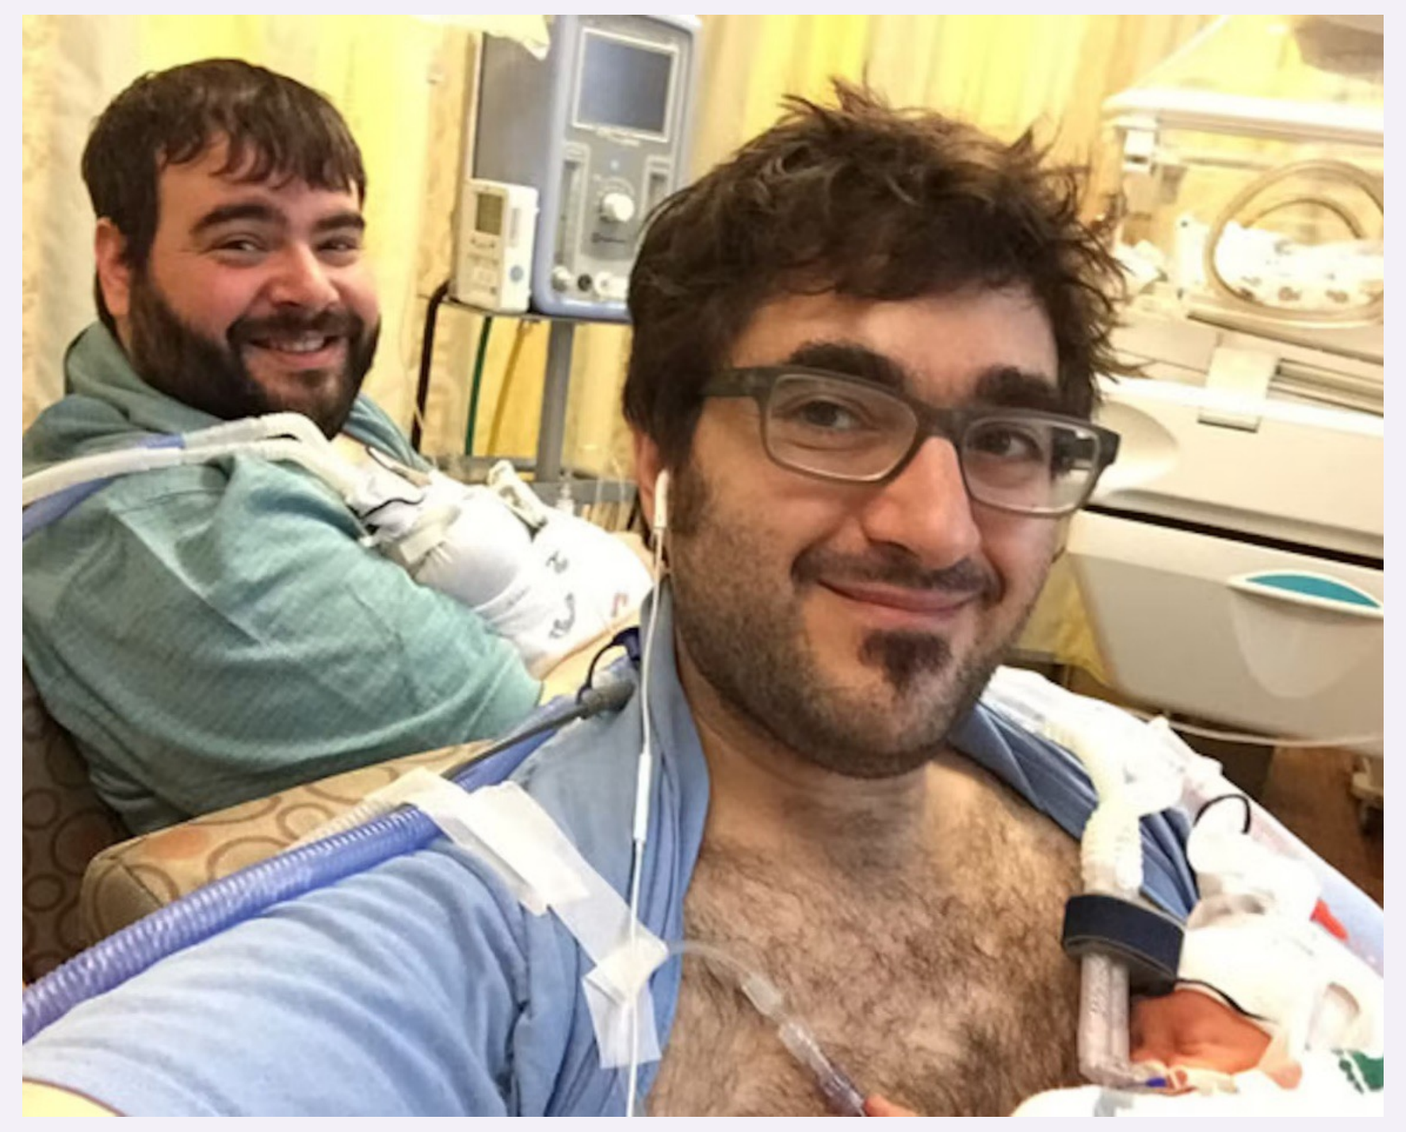

Photo Source: A two-dad family's journey through the NICU (Facebook)

Application of results to theoretical framework

NICU nurses' comprehensibility of partnering with fathers and non-birth parents in family-centered care in the NICU

- Nurses acknowledge that family-centered care is important
- Nurses' hesitation to include parents in family-centered care
 - Parents seen as hindrance; infant safety
- Birth mother is prioritized over father or non-birth parent
 - Experienced pregnancy, childbirth; hormones; breastfeeding
- Fathers' apprehension about participating in care
 - Fear of harming infant; discomfort with NICU environment
 - Sometimes fathers defer to mothers; need encouragement
- Same-sex and/or adoptive parents treated the same as traditional parents

NICU nurses' manageability related to partnering with fathers and non-birth parents in family-centered care in the NICU

- Fathers not as present at bedside as mother
 - Work responsibilities; caring for other children; social situation
- Fathers' apprehension to participate in care
 - Fragility of infant; uncomfortable in NICU environment
- Same-sex or adoptive parents treated same as traditional parents

Meaningfulness of NICU nurses related to partnering with fathers and non-birth parents in family-centered care in the NICU

- Family-centered care is important
 - Helping parents gain confidence in caring for their child is meaningful
 - Satisfaction from preparing parents for discharge
 - One participant conveyed, "It's fun to teach them"
- Enjoys helping them hold their infant for the first time

Research support for this investigation was provided by the Yvonne L. Munn Predoctoral Research Grant for PhD Nursing Students and the Yvonne L. Munn Center for Nursing Research, Institute for Patient Care, Massachusetts General Hospital